2252

Stabilized decompositions for improved cardiac self-gating: A proof-of-concept in single breath-hold 3D cine imaging

Anna Padée[1], Lorenzo Di Sopra[2], John Heerfordt[2,3], Jérôme Yerly[2,4], Marco Merlo[1], Tobias Kober[2,3,5], Davide Piccini[2,3,5], Matthias Stuber[2,4], Christopher Roy[2], and Jonas Richiardi[2,3]

[1]Laboratory for Psychiatric Neuroscience and Psychotherapy, University of Fribourg, Fribourg, Switzerland, [2]Department of Radiology, Lausanne University Hospital and University of Lausanne, Lausanne, Switzerland, [3]Advanced Clinical Imaging Technology, Siemens Healthcare, Lausanne, Switzerland, [4]Center for Biomedical Imaging, Lausanne, Switzerland, [5]LTS5, Ecole Polytechnique Fédérale de Lausanne, Lausanne, Switzerland

Synopsis

ECG recording during whole heart MR imaging requires extra setup time and is not always reliable. Recently, self-gated (SG) acquisition techniques have been developed, which extract the cardiac signal from k-space center amplitude modulations. Here, we investigated whether different single- (Empirical Mode Decomposition (EMD), Ensemble EMD (EEMD)) and multi-coil (PCA, ICA with novel stabilisation) decomposition methods yielded SG triggers with minimal variability to ECG R-wave location. EEMD yielded median variability similar to PCA. Compared to PCA, our proposed stabilised ICA approach yielded SG triggers with 66% lower variability (median over 5 subjects), although not for all subjects.

INTRODUCTION:

Motion-resolved imaging[1,2] of the heart depends on reliable physiological gating signals. ECG-triggering is commonly used for synchronising the MR acquisition to the cardiac cycle. Recently, self-gating (SG) methods have shown promise for simplifying the setup by instead extracting the cardiac motion information directly from the acquired imaging data[3]. Approaches for extracting self-gating signals have mainly rely on principal component analysis (PCA)[1,2,4], or independent component analysis (ICA)[1]. However, PCA and ICA assume linearity and stationarity of the analysed time-series, which may not be realistic in cardiac imaging due to e.g. non-sinus rhythm, and ICA runs can yield very different components[5]. Therefore, we hypothesize that decomposition methods with less restrictive stationarity assumptions or more stability may result in improved cardiac SG signal extraction. Given varying coil distances to the myocardium, it is also uncertain whether all coils are needed to recover an SG signal, and therefore we explored both univariate methods (single-coil) and multivariate methods (multi-coils).

METHODS:

Datasets:
Single breath-hold (scan time=28s) 3D cine datasets were acquired in N=5 healthy volunteers on a 1.5T clinical MRI scanner (MAGNETOM Aera, Siemens Healthcare) using a free-running 3D golden angle radial bSSFP[6,7] sequence. An ECG was simultaneously recorded for analysis purposes. The signals of interest were obtained by selecting the absolute value of the central coefficient of periodically acquired k-space readouts (K0 modulation).
Decomposition approaches:
We tested single-coil and multi-coil decompositions. Single-coil decomposition were performed on all coils sequentially. Empirical mode decomposition (EMD), a single-coil method, is particularly suited for non-stationary signals, but can be affected by mode mixing. Thus, we also used ensemble EMD (EEMD), which improves over EMD by running the decomposition multiple times with added white noise, turning EMD into an adaptive dyadic filter bank. Averaging over the runs makes the method less sensitive to the perturbations to data[8]. For multi-coil decompositions, we used the ICASSO principle[5] with FastICA[9] implementation to obtain 12 components over 200 runs of ICA. We then clustered all components into 12 clusters (Figure 3) using hierarchical clustering with a Ward variance minimization algorithm.

We propose two automated approaches to picking a decomposition from the multiple ICA runs. In the centrotype ICA approach, we estimated the sources by averaging all components within each cluster. This centrotype was then used to compute “pseudo sources” from the original signals, with no independence guarantee. In the proposed second approach, stabilised ICA (StabICA), we selected the most reliable decomposition based on a cluster quality index I_q: the difference between average within-cluster(C_m) similarities $\sigma_{i,j}$ and average between-cluster(C_{-m}) similarities:

$$I_q(C_m) = \frac{1}{|C_m|^2} \sum_{i,j \in C_m} \sigma_{i,j} - \frac{1}{|C_m||C_{-m}|} \sum_{i \in C_m} \sum_{j \in C_{-m}} \sigma_{i,j}; \text{where } C_{-m} = C - C_m$$

These methods were compared to PCA as a baseline.
Automatic SG component selection:
For each dataset, the subject-specific heartrate was extracted using FFT as the frequency with maximum power in the 0.5-2Hz band. This frequency was later used to automatically identify the components of interest, by selecting the component with the highest power spectral density in the ±0.3Hz band around heart rate.
Evaluation metrics:
The timing differences between peaks of the SG signal and triggers obtained from ECG R-waves were computed for each of the around 25 heartbeats per dataset. The distance between SG peaks varies, due to delay between the electrical activity and heart motion, ECG filtering and sign indeterminacy of ICA. Thus, we used standard deviation of the distribution of timing differences as the evaluation measure. The distributions of standard deviations were compared subjectwise between methods using the robust Bonett-Seier test of scale for paired samples[10].

RESULTS:

Most methods were able to recover a component representing the cardiac cycle. Specifically, StabICA, Centrotypes ICA, PCA, and EEMD recovered all triggers in 4 out of 5 subjects, while EMD recovered all triggers only in 2 cases. No method recovered all triggers for subject 4 (see Figure 3). The median standard deviation of the differences between SG triggers and ECG triggers across the 5 datasets were: StabICA: 13.2 ms, centrotypes: 12.5 ms, PCA: 37.8 ms, EMD: 61.3ms and EEMD: 36.9 ms. Figure 2 shows standard deviations for the various decomposition of one dataset. Two subjects had significantly lower variability with StabICA than PCA (p=0.005, p<10-16), one had lower variability but no significant difference (p=0.25), and two had higher variability with StabICA. Figure 3 shows results for all subjects. Qualitative examination of ECG, PCA, and StabICA reconstruction for one subject showed no visible quality difference (Figure 5).

DISCUSSION/CONCLUSION:

Stabilized ICA and Centrotypes ICA showed lower variability than other decomposition methods. This supports our hypothesis that fewer assumptions might provide more “ECG-like” cardiac self-gating, as gating based on the ECG signal itself has no such assumption. However, the ECG as a reference standard is not ideal due to missed triggers or falsely identified triggers caused by MHD effects. More generally, our proposed approaches could improve over vector gating, which requires manual selection of the ECG vector and constant monitoring for missed triggers. Image quality on an example subject was not qualitatively different between SG decomposition methods, and future work includes expanding the analysis to more subjects including patients with arrhythmia, varying sequences, as well as solving sign indeterminacy of decompositions to further improve consistency.

Acknowledgements

No acknowledgement found.

References

1.Di Sopra L, et al. An automated approach to fully self-gated free-running cardiac and respiratory motion-resolved 5D whole-heart MRI. Magn. Reson. Med. 2019;82:2118–2132

2. Feng L, et al. 5D whole-heart sparse MRI. Magn. Reson. Med. 2018;79:826–838 doi: 10.1002/mrm.26745.

3. Larson A.C, et al. Self-Gated Cardiac Cine MRI Magnetic Resonance in Medicine 51:93–102 (2004)

4. Pang J, et al. ECG and navigator-free four-dimensional whole-heart coronary MRA for simultaneous visualization of cardiac anatomy and function. Magn. Reson. Med. 2014;72:1208–1217 doi: 10.1002/mrm.25450.

5. Himberg J, Hyvärinen A. Icasso: software for investigating the reliability of ICA estimates by clustering and visualization. In Proc. 2003 IEEE Workshop on Neural Networks for Signal Processing (NNSP2003), pp. 259-268, Toulouse, France, 2003

6. Piccini D, et al. Four-dimensional whole-heart cine imaging with isotropic resolution in one single breath-hold: initial results. Proc. ISMRM 2019.

7. Coppo S, et al. Free-running 4D whole-heart self-navigated golden angle MRI: Initial results. MRM 2014

8. Zhaohua W, Norden E. Ensemble Empirical Mode Decomposition: a Noise-Assisted Data Analysis Method Advances in Adaptive Data Analysis, Vol. 1, No. 1 (2009)

9. Hyvärinen A, Oja E. Independent Component Analysis: Algorithms and Applications, Neural Networks, 13(4-5):411-430, 2000

10. Bonett D.G, Seier E. Statistical inference for a ratio of dispersions using paired samples. Journal of Educational and Behavioral Statistics, 28, 21-30, 2003

Figures

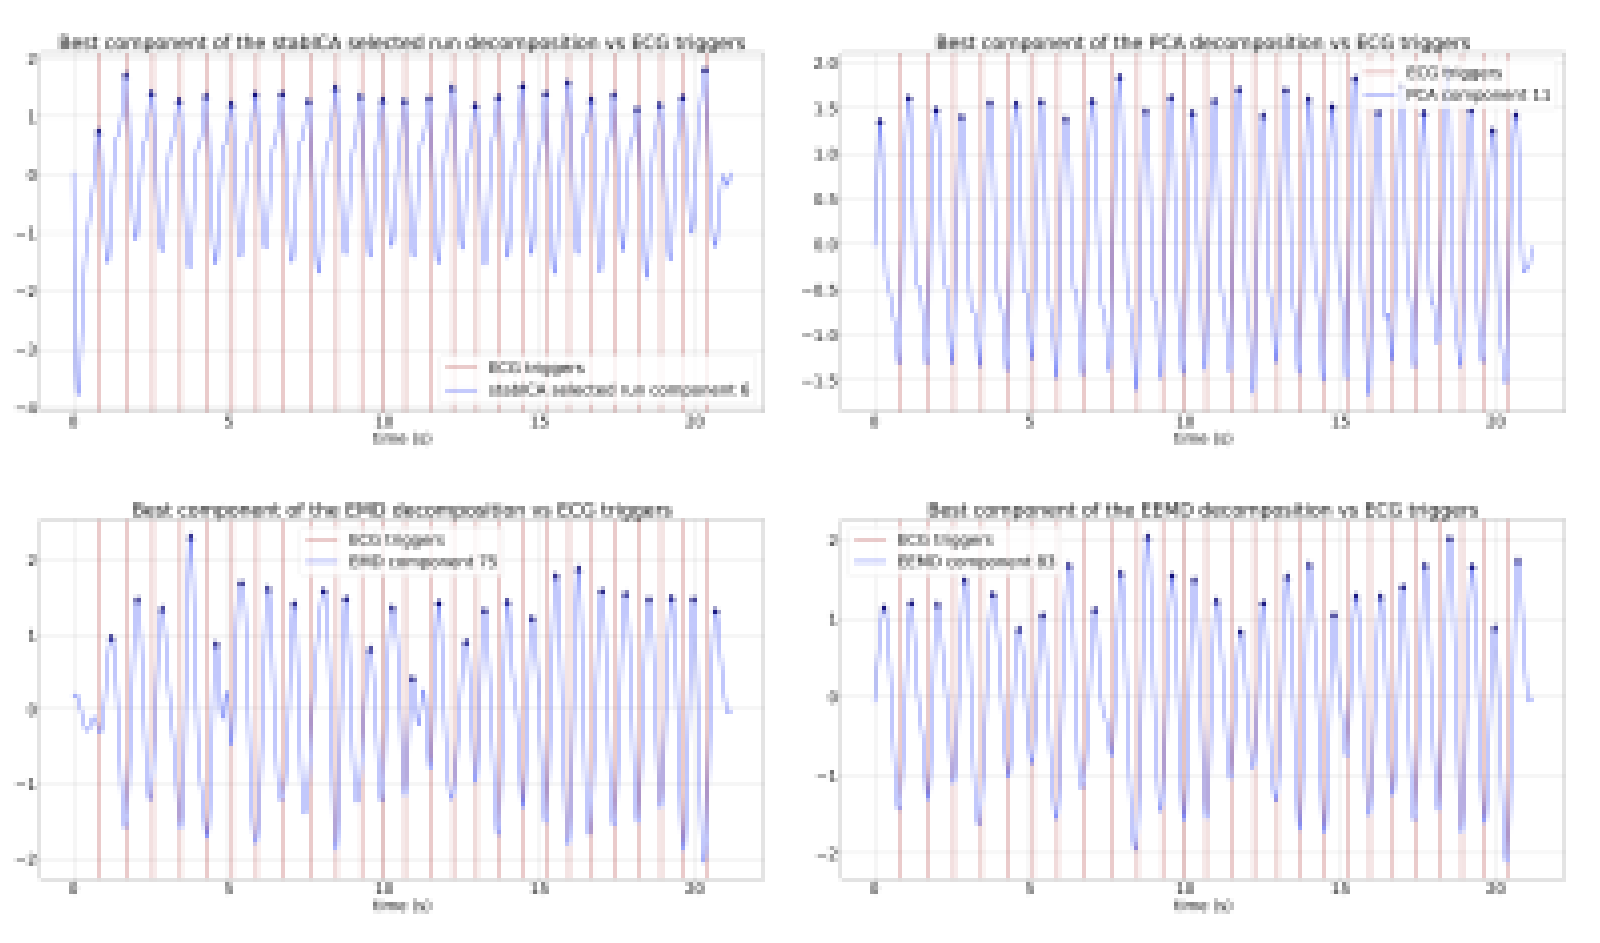

Example decompositions for one dataset (subject 1). For each of the decomposition methods, one component of interest was extracted, based on the previously established heart rate. The extracted component was then band-pass filtered to remove any high-frequency noise, yielding an SG signal estimate.

Component of interest for: (a) stabilised ICA (b) PCA (c) EMD (d) EEMD. Vertical lines mark ECG triggers (reference), blue lines show components with triangles showing the estimated SG triggers. EMD displays mode mixing (c), while EEMD largely eliminates it (d)

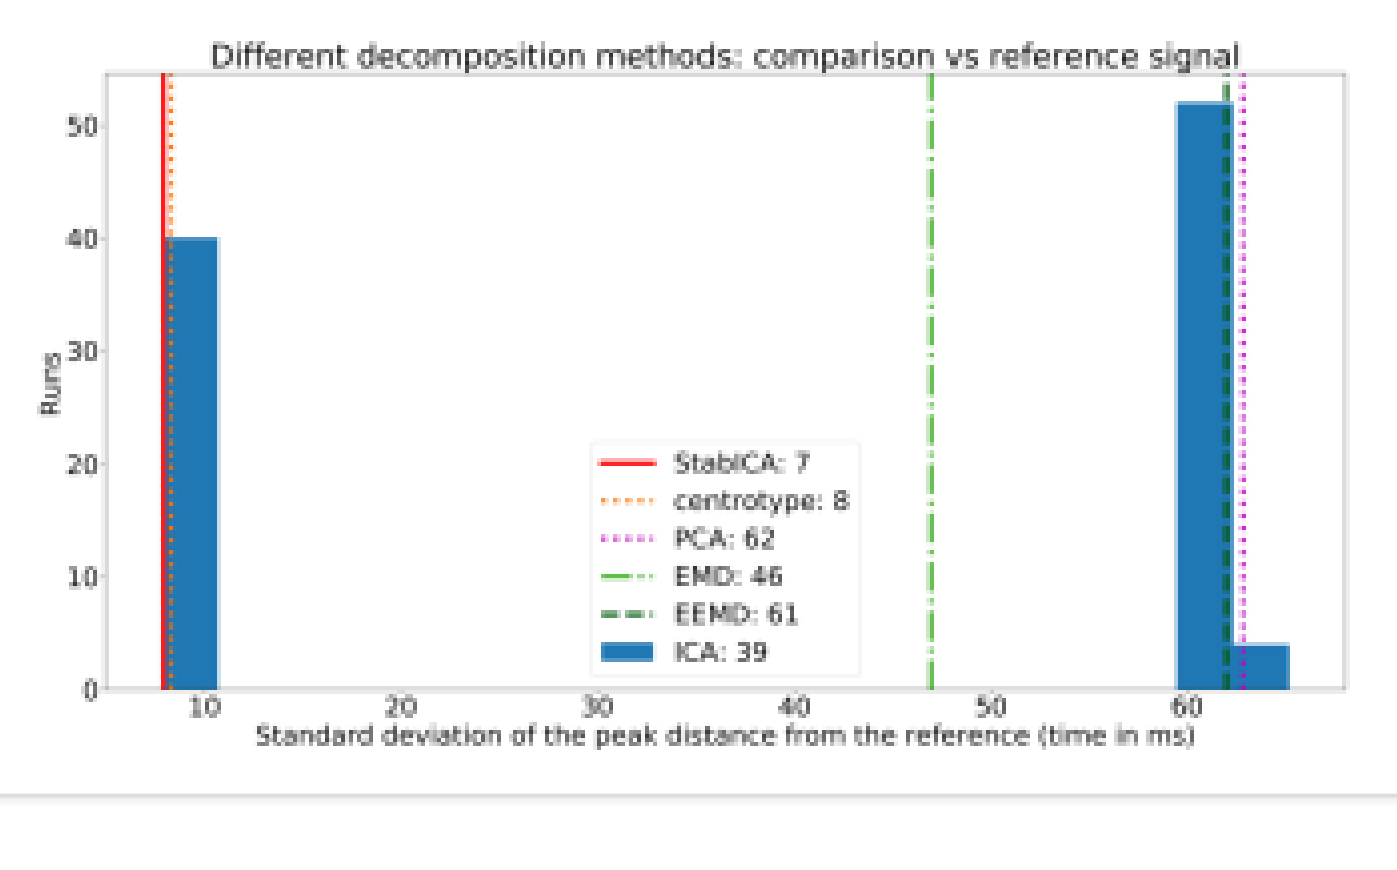
Different decomposition methods: comparison vs reference signal
Runs
Standard deviation of the peak distance from the reference (time in ms)
StabICA: 7
centrotype: 8
PCA: 62
EMD: 46
EEMD: 61
ICA: 39

Comparison of standard deviations with respect to ECG R-wave trigger for different decompositions on one dataset. Each decomposition's automatically selected best SG component is shown by vertical lines. To illustrate variability between ICA runs and motivate the use of stabilised ICA or centrotypes ICA, a histogram of standard deviations for SG trigger obtained from the best component of each ICA run is shown in blue. Lower standard deviation represents SG components with more consistent timing with respect to the ECG reference. Non-converged ICA runs are not shown.

Subject	StabICA	PCA	Centrotypes	EMD	EEMD
1	13.2	50.0	12.5	39.3	32.9
2	7.9	8.7	48.1	39.4	36.9
3	17.5	13.0	7.7	61.7	19.7
4	261.1	37.8	256.7	67.4	136.2
5	10.3	82.0	10.8	61.3	80.9

Standard deviations [ms] across cardiac cycles for timing between SG triggers provided by different decomposition and ECG R-wave trigger. Visual inspection shows that the bad performance of StabICA for subject 4 is probably due to sign indeterminacy of the chosen component.

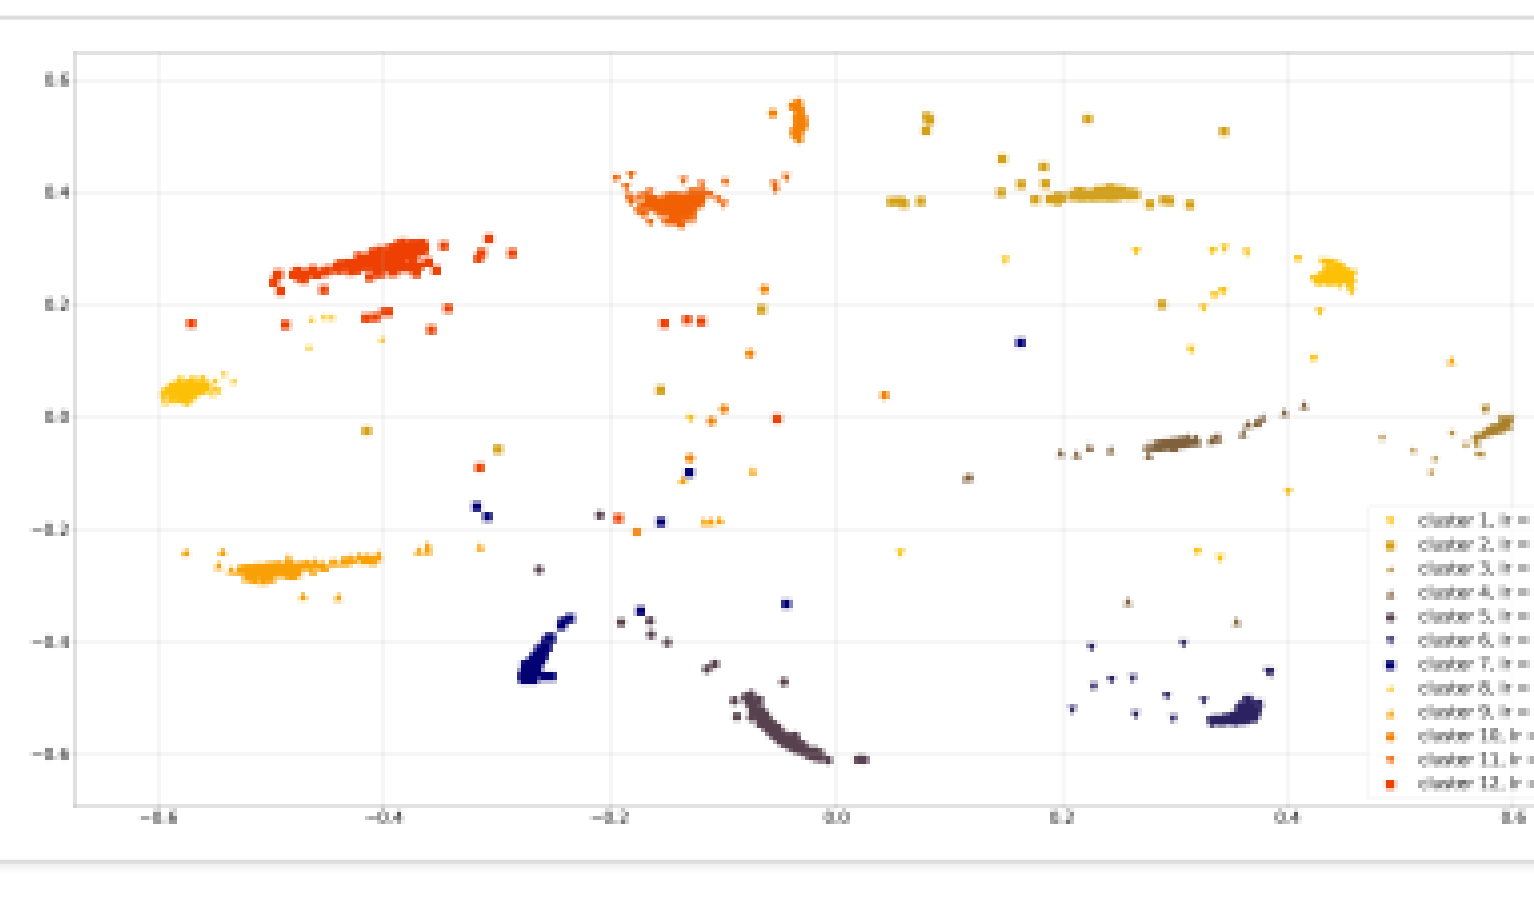

Clustering of ICA runs. 2D multidimensional scaling projection of clustering of all the components from 350 FastICA runs with the cluster quality index I_r. Components were aggregated into 12 clusters based on their similarities. FastICA is not deterministic due to initialization, finite sample sizes and local minima which imply that results vary from run to run, and the reliability of components is unknown.Tighter and better separated clusters correspond to stable components.

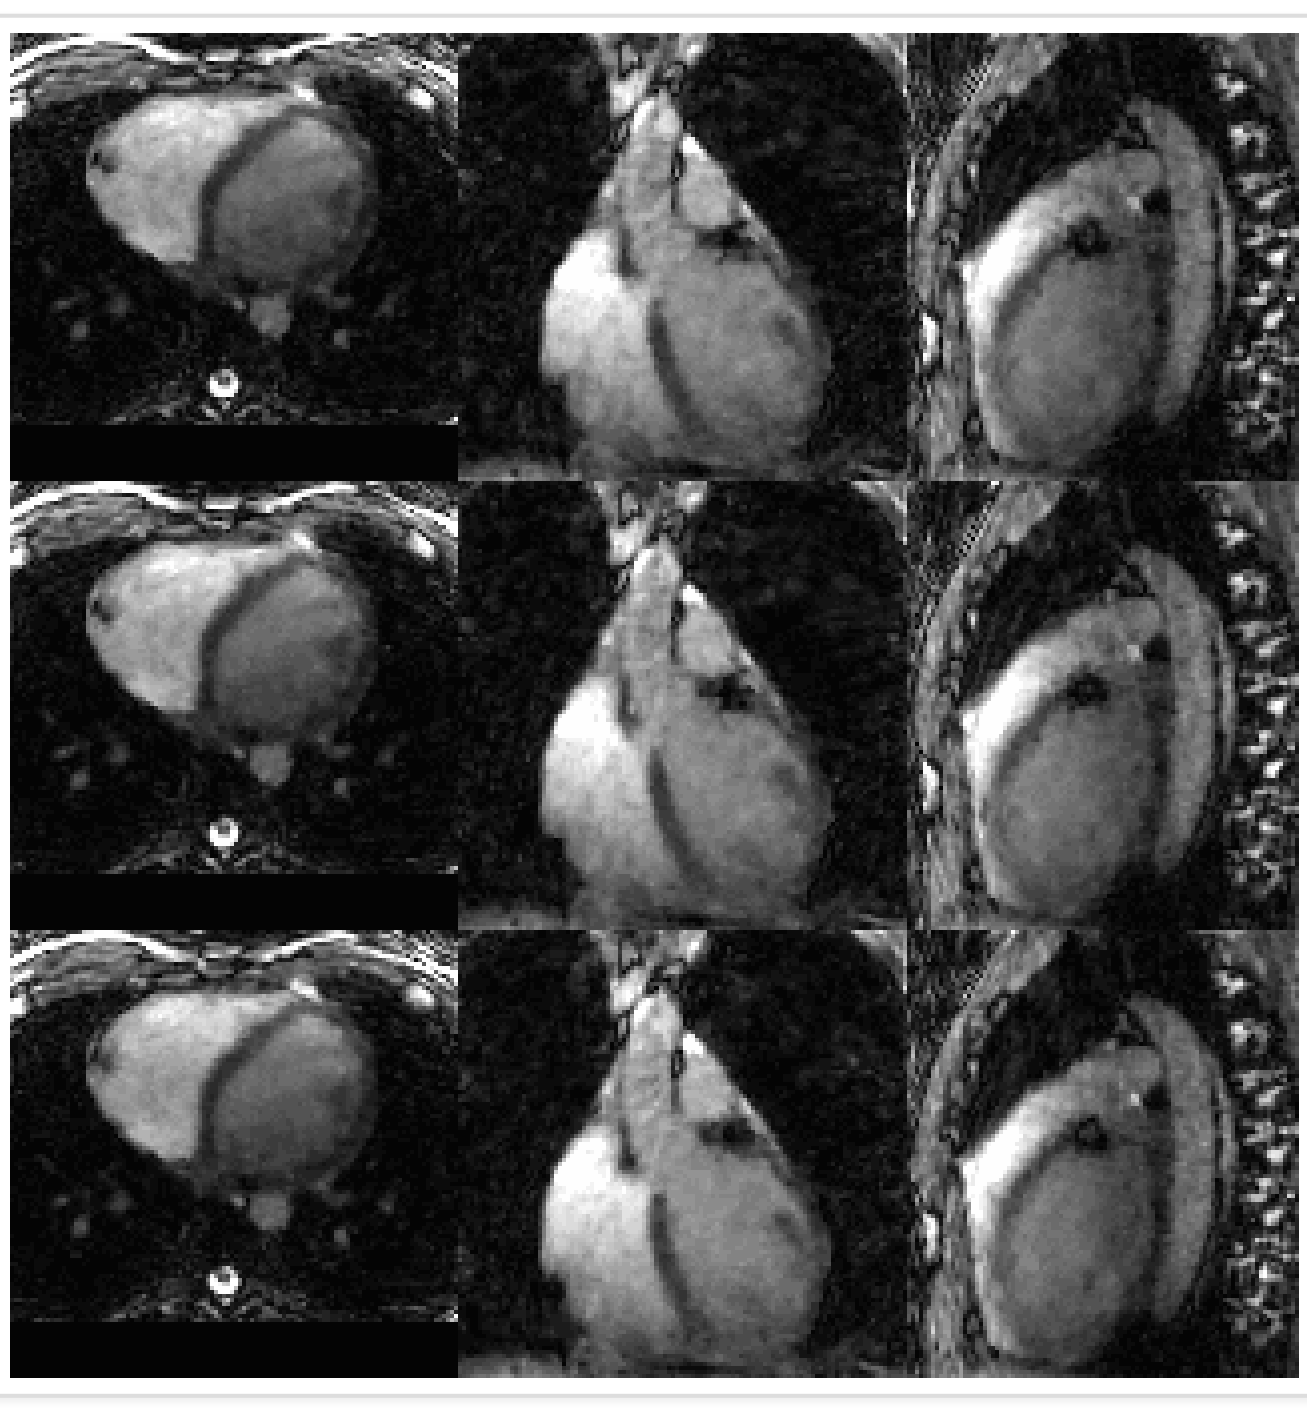

Reconstructions for subject 2 from the triggers recovered by StabICA(top), worst ICA run(middle), and ECG(bottom) reference. To show the impact of ICA variability on image quality, we compared reconstructed images for triggers extracted with stabilized ICA and the worst of all computed ICA decompositions. The worst decomposition was chosen as the one with the highest standard deviation of the difference from ECG triggers. A single ICA run has a risk of producing a non-optimal trigger recovery, which can impact image quality

Proc. Intl. Soc. Mag. Reson. Med. 28 (2020) 2252